Molecular Locomotion: Modeling the Functional Domains of E1 Helicase to Assess the Asymmetric Rotary Model

Authors: Lily Bailey, Nora Bradford, Simon Chigier, John Gustafson, Hansika Iyer, Michael Liapin, McLean Sherrin, Bowen Zhan, Judith Rokous Mentor: Ernst ter Haar; Company: Vertex Pharmaceuticals

SMART Teams 2014-2015 MSOE University

Abstract: Helicases are a highly conserved class of enzymes that unwind the double helix of DNA, providing access to single-stranded DNA. Found in all living organisms and viruses, helicases function as motor proteins in replication, transcription, and remodeling of DNA. A common sexually-transmitted oncovirus, human papillomavirus (HPV) uses E1 helicase, the most studied helicase protein because of the ease by which it can be isolated. The only enzyme encoded in the HPV episome, E1 hexameric helicase is a ring-shaped translocase that belongs to the AAA+ family and superfamily 3. E1 has near exact rotational symmetry with respect to its six subunits. Each monomer comprises a central five-stranded anti-parallel beta-sheet and six alpha helices. Conserved sequence motifs Walker A and B and an arginine finger participate in the binding and hydrolysis of ATP causing conformational changes in β-hairpins, powering the walking movement along the ssDNA. Assembling first as a double-trimer at the replication origin and then into two homohexamers on opposing strands, E1 separates the dsDNA into ssDNA by occlusion in the 3’→5’ direction. Asymmetric sequential progression of ATP hydrolysis around the hexameric ring at catalytic sites between monomers powers translocation of E1. An understanding of its functional domains and how they interact with each other, DNA, and host proteins is necessary to elucidate the overall mechanism of helicase function. Using 3D printing technology, The Governor’s Academy SMART (Students Modeling A Research Topic) Team modeled the E1 helicase to provide vital insight into the mechanism of eukaryotic DNA replication and potentially promote development of therapeutic treatments.

HPV Viral Life Cycle

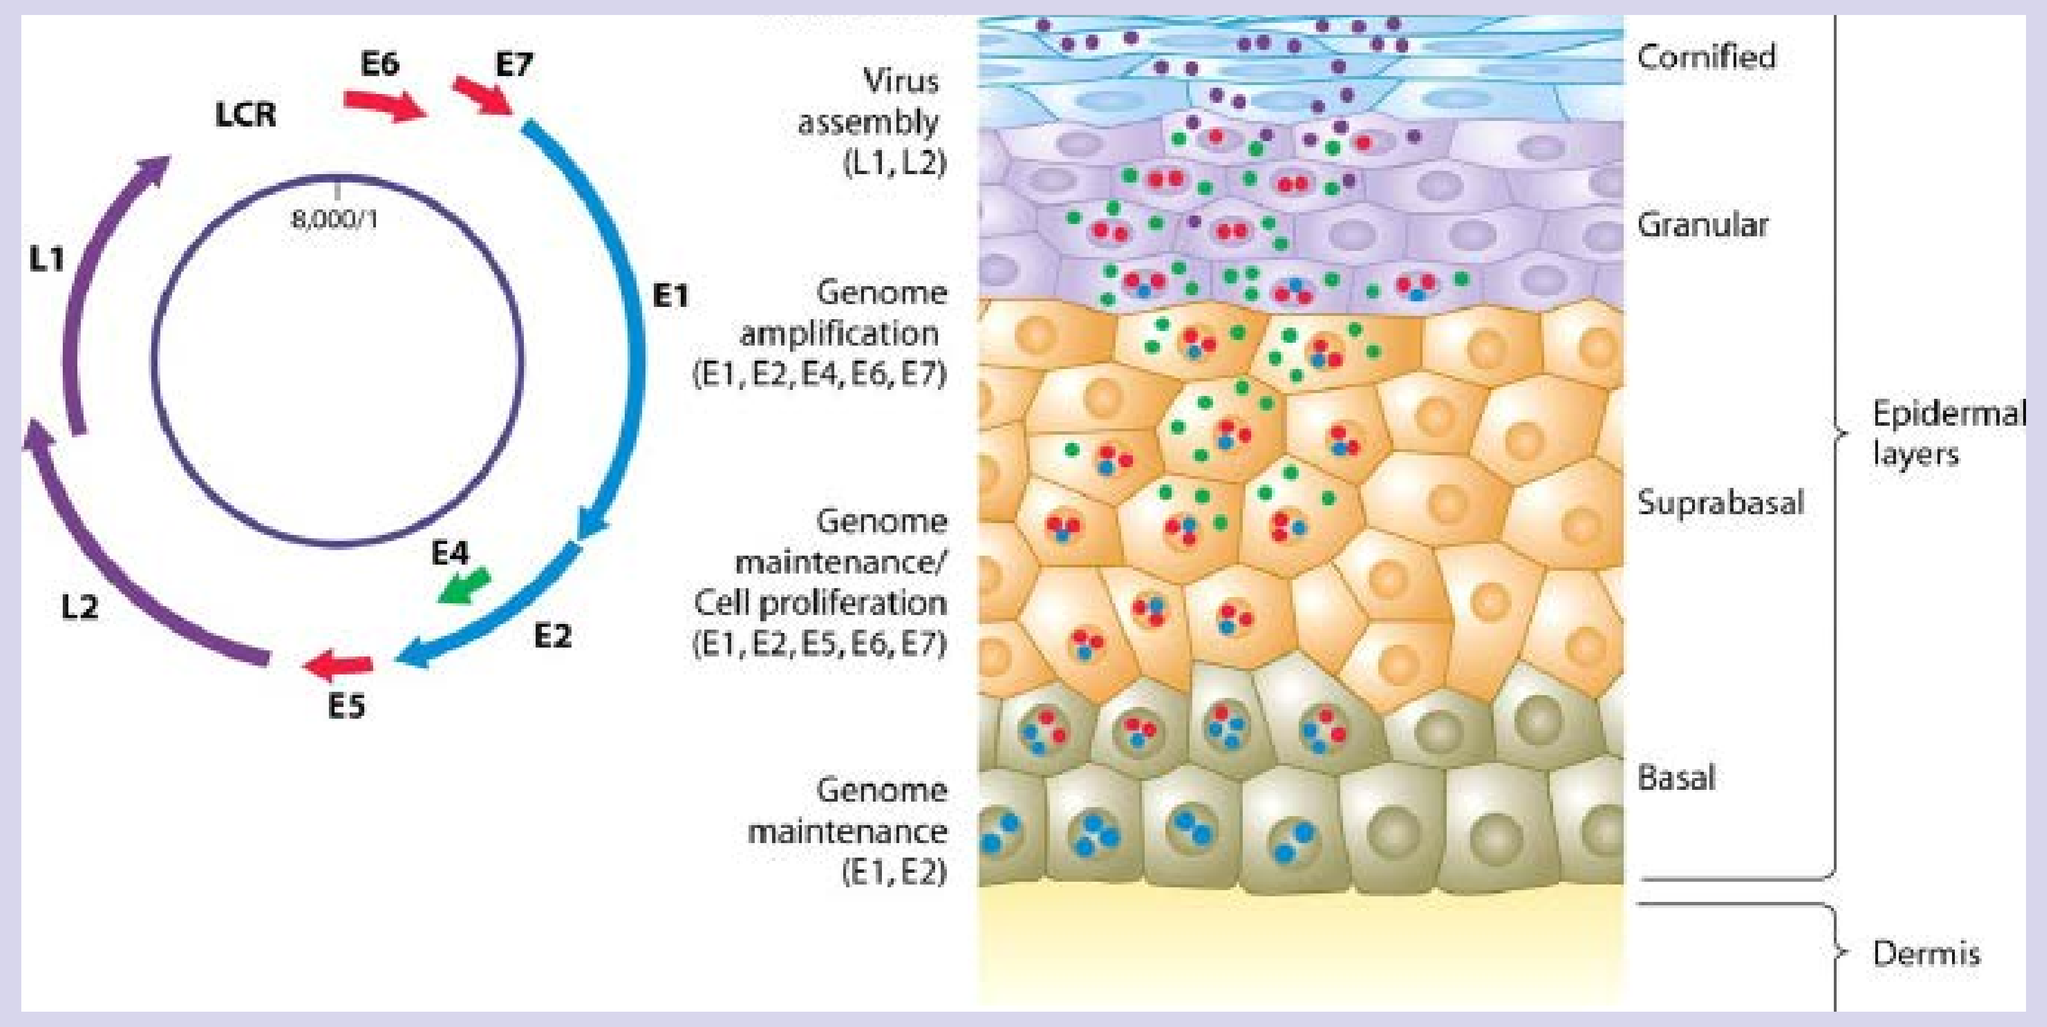
LCR
E6
E7
E1
E2
E4
E5
L1
L2
8,000/1
Virus assembly (L1, L2)
Genome amplification (E1, E2, E4, E6, E7)
Genome maintenance/ Cell proliferation (E1, E2, E5, E6, E7)
Genome maintenance (E1, E2)
Cornified
Granular
Suprabasal
Basal
Epidermal layers
Dermis

Virus infects cells via microabrasions; host cell expresses viral proteins; unscheduled cell division & viral genome amplification; virus & infected cells mature & move up layers

Assembly of E1 at the origin of replication

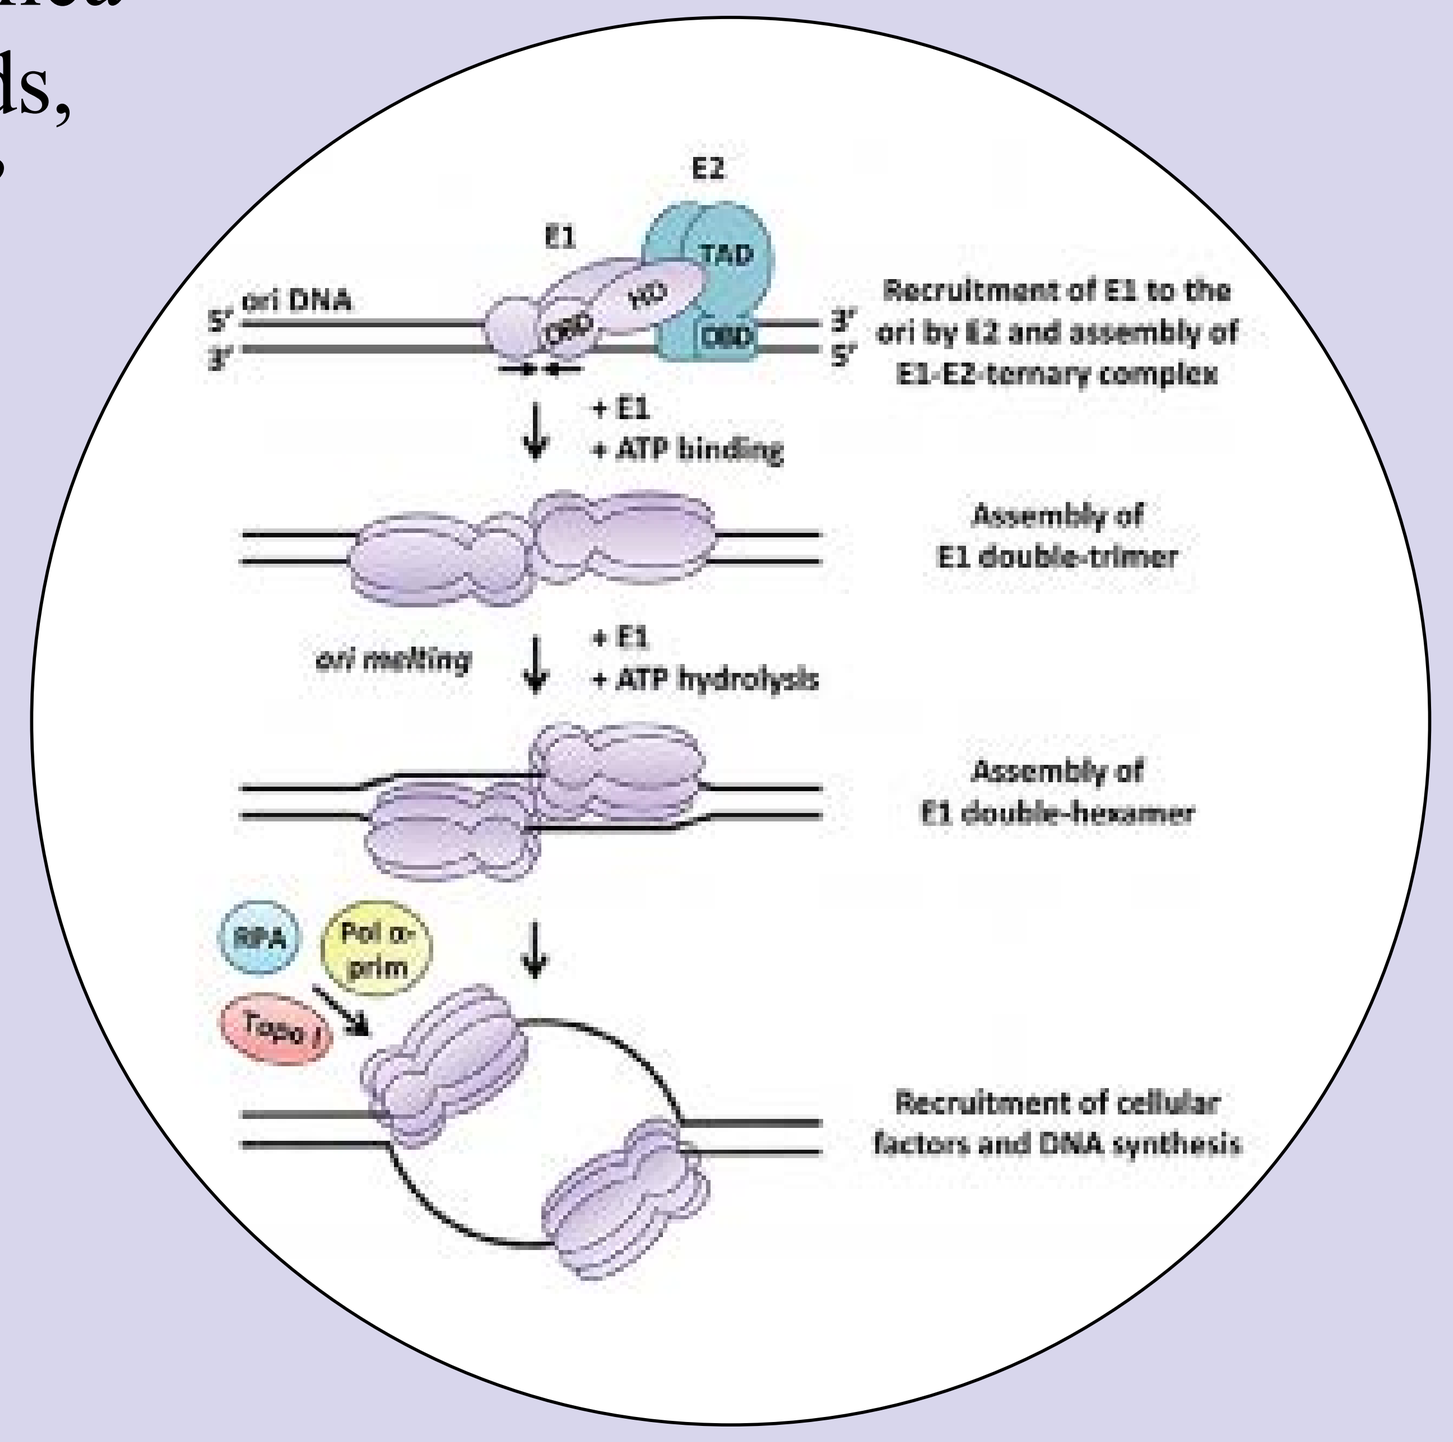
E2
E1
TAD
ori DNA
Recruitment of E1 to the ori by E2 and assembly of E1-E2-ternary complex
+ E1
+ ATP binding
Assembly of E1 double-trimer
ori melting
+ E1
+ ATP hydrolysis
Assembly of E1 double-hexamer
RPA
Pol α-prim
Topo I
Recruitment of cellular factors and DNA synthesis

Introduction: Human papillomavirus (HPV) is a sexually transmitted DNA oncovirus that causes genital warts and cervical cancer. Upon infection of basal skin stem cells, HPV proteins cause unscheduled cell division forming lesions from the basal layer up. As cells move up through layers of the epidermis, they differentiate to a terminal state in the outer layer where the mature virus leaves. E1 hexameric helicase plays an essential role during the viral life cycle and is the only enzyme encoded by both HPV and closely-related bovine papillomavirus (BPV). Mutant forms of the enzyme are associated with cancer and premature aging. Serving as a molecular motor protein vital to unwinding DNA, E1 is a member of superfamily 3, and the AAA+family (ATPases associated with cellular activities) which are characterized by their conserved nucleotide-binding module. It is one of the most well-characterized helicases because it is easily isolated and produces reliable x-ray crystallography results. E1 assembly begins with the formation of two initial trimers which bind to the origin of replication (with aid from E2), where they assemble to form a double hexamer. Each subunit comprises five α-helices and four β-sheets. The six subunits assemble to form a ring-shaped hexamer with beta hairpins protruding into a narrow 13-20 Å central channel that can only accommodate ssDNA. At each interface between monomers, ATP-binding sites are created by conserved Walker A, Walker B, and sensor 1 motifs on one monomer, and sensor 2, sensor 3, and an arginine finger on the adjacent monomer. E1 helicase most likely changes conformation via an “asymmetric rotary” mechanism by which the six subunits cycle between ATP, dephosphorylated ATP, and apo (or empty) states.

Replisome; hexamers move in opposite directions 3’ to 5’

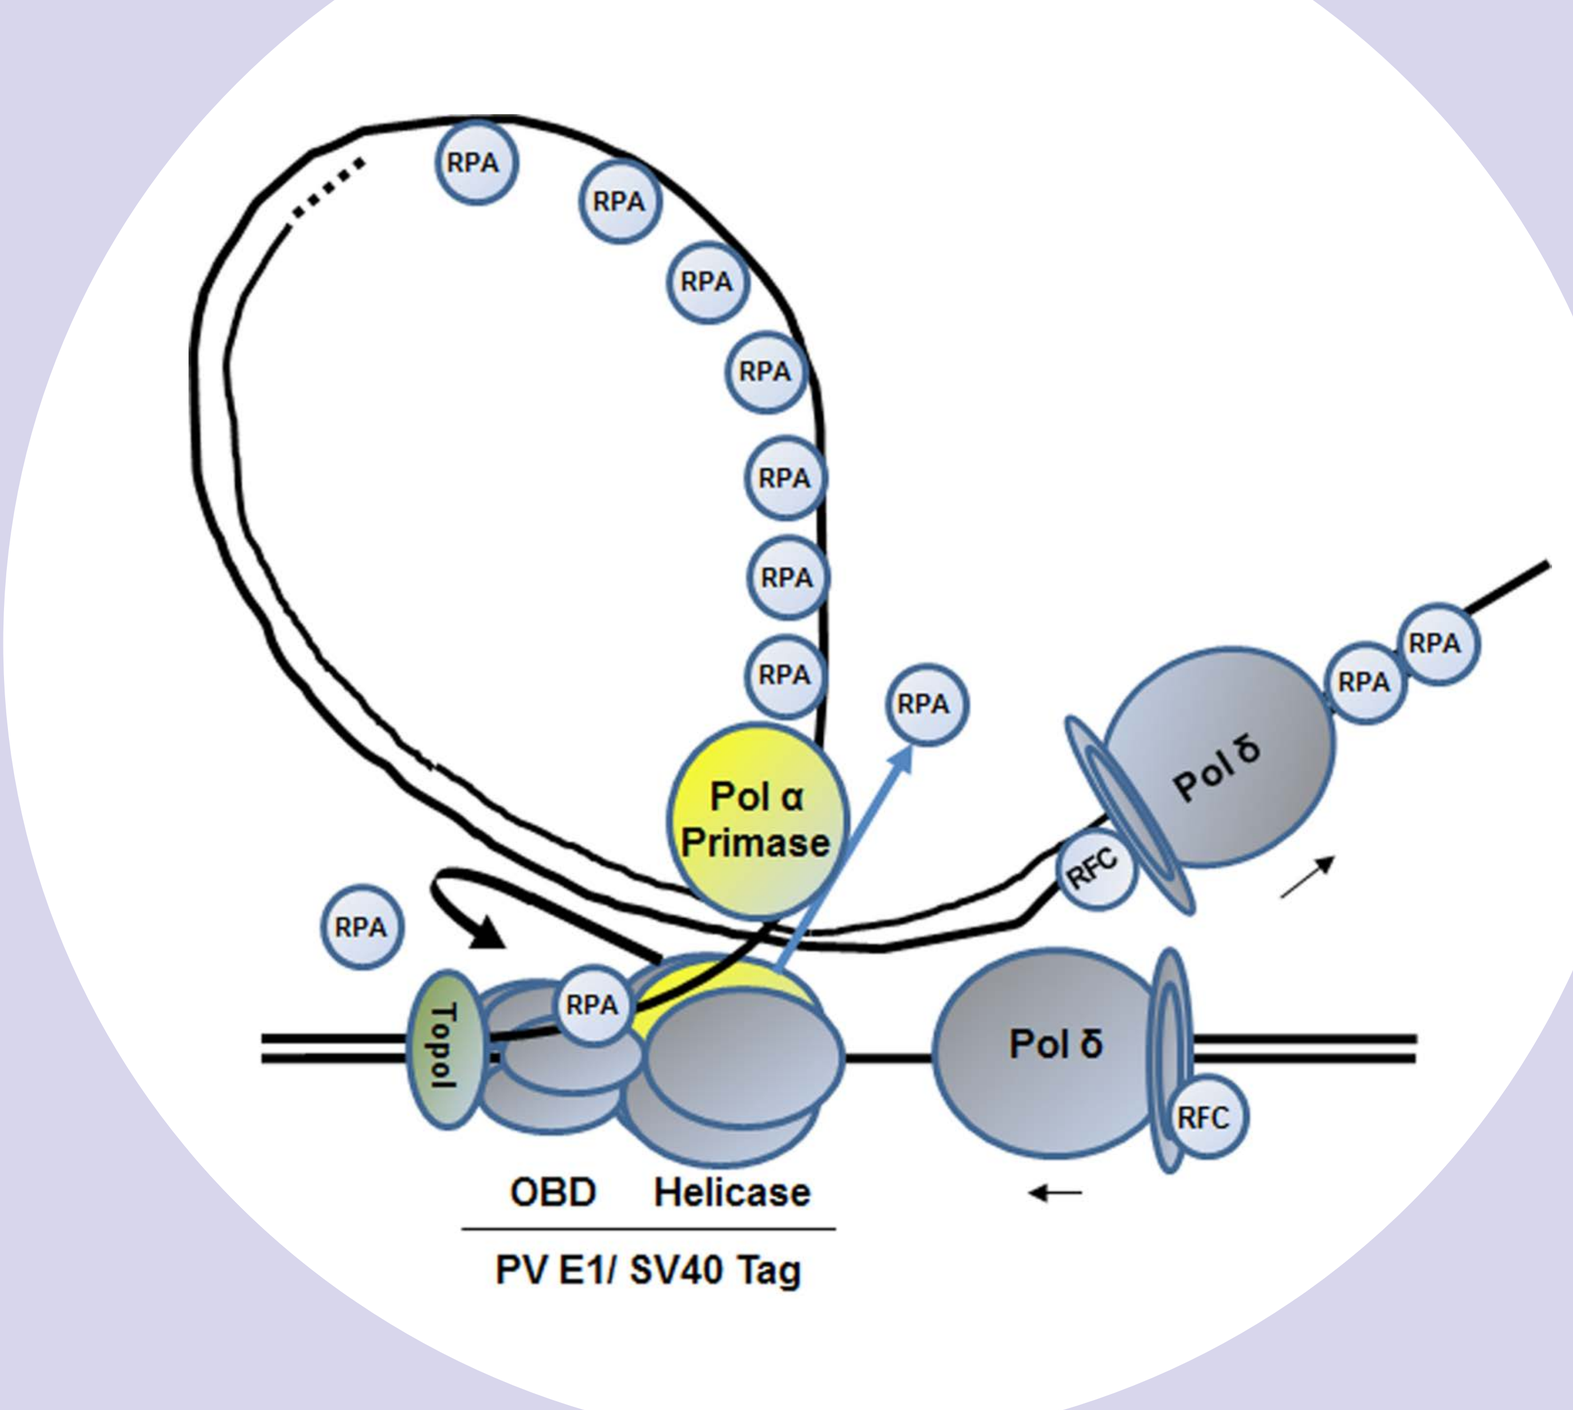
RPA
Pol α Primase
Pol δ
RFC
TopoI
OBD
Helicase
PV E1/ SV40 Tag

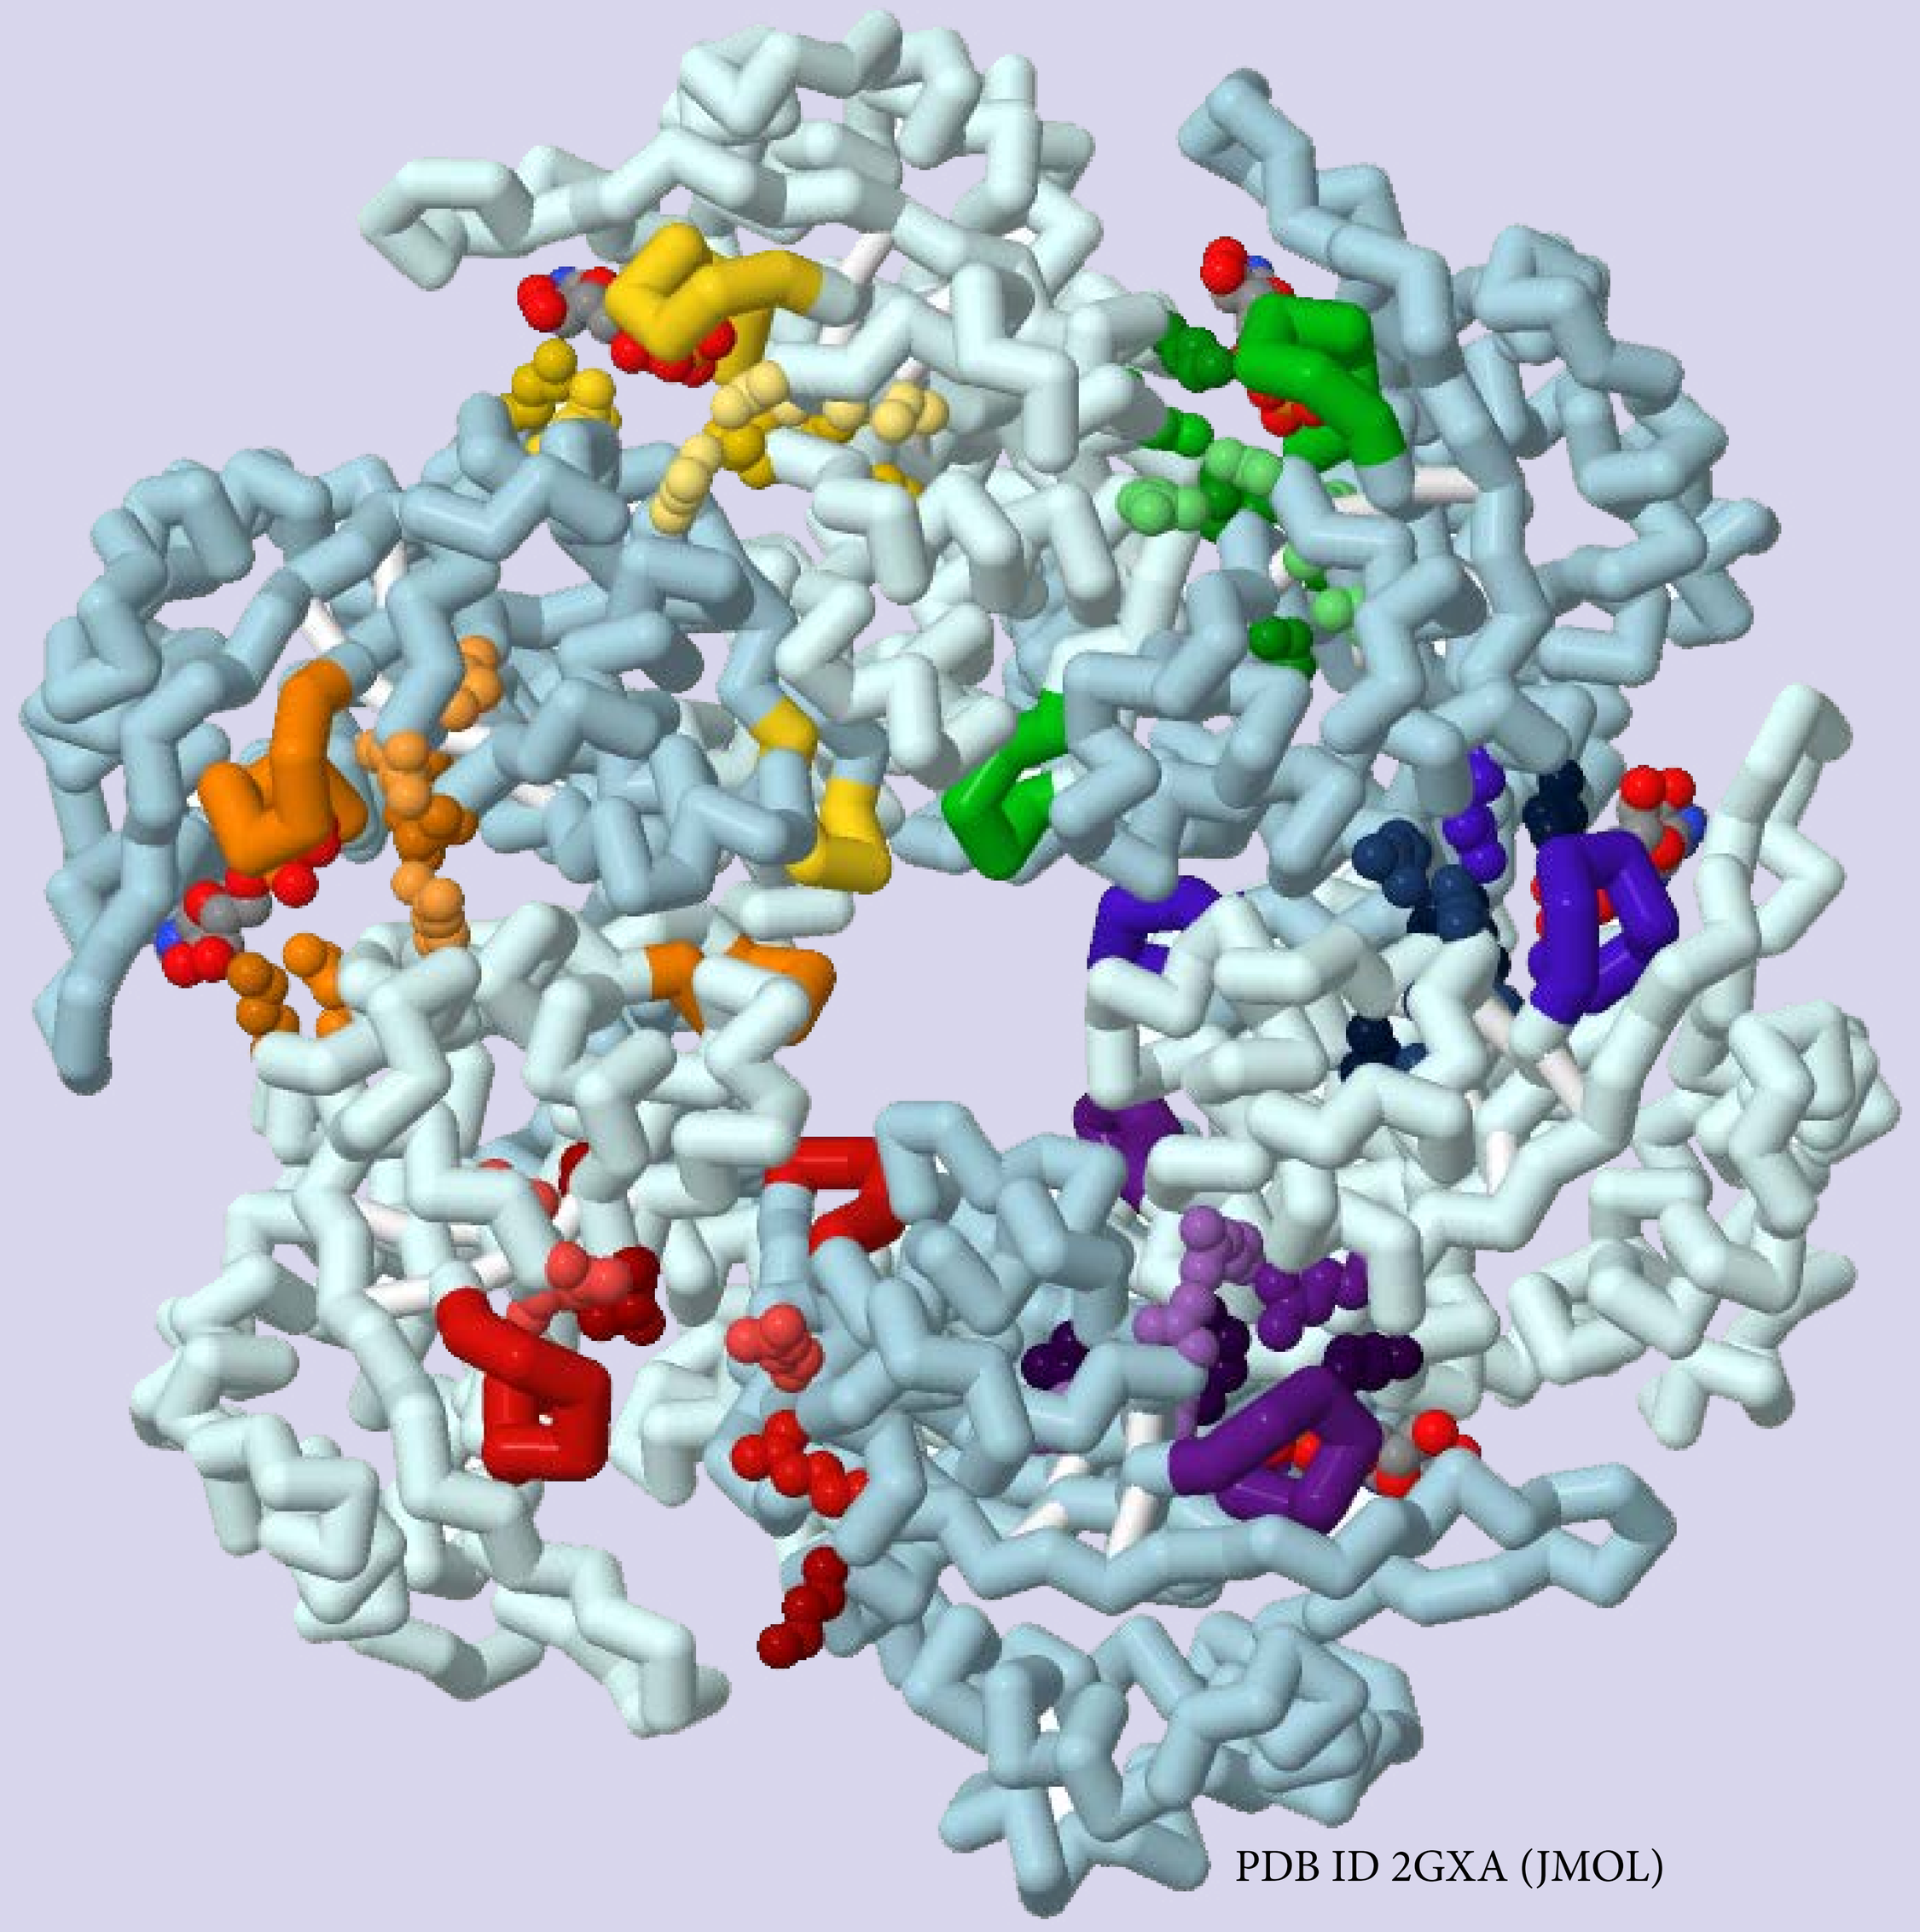

PDB ID 2GXA (JMOL)

Domain structure of E1

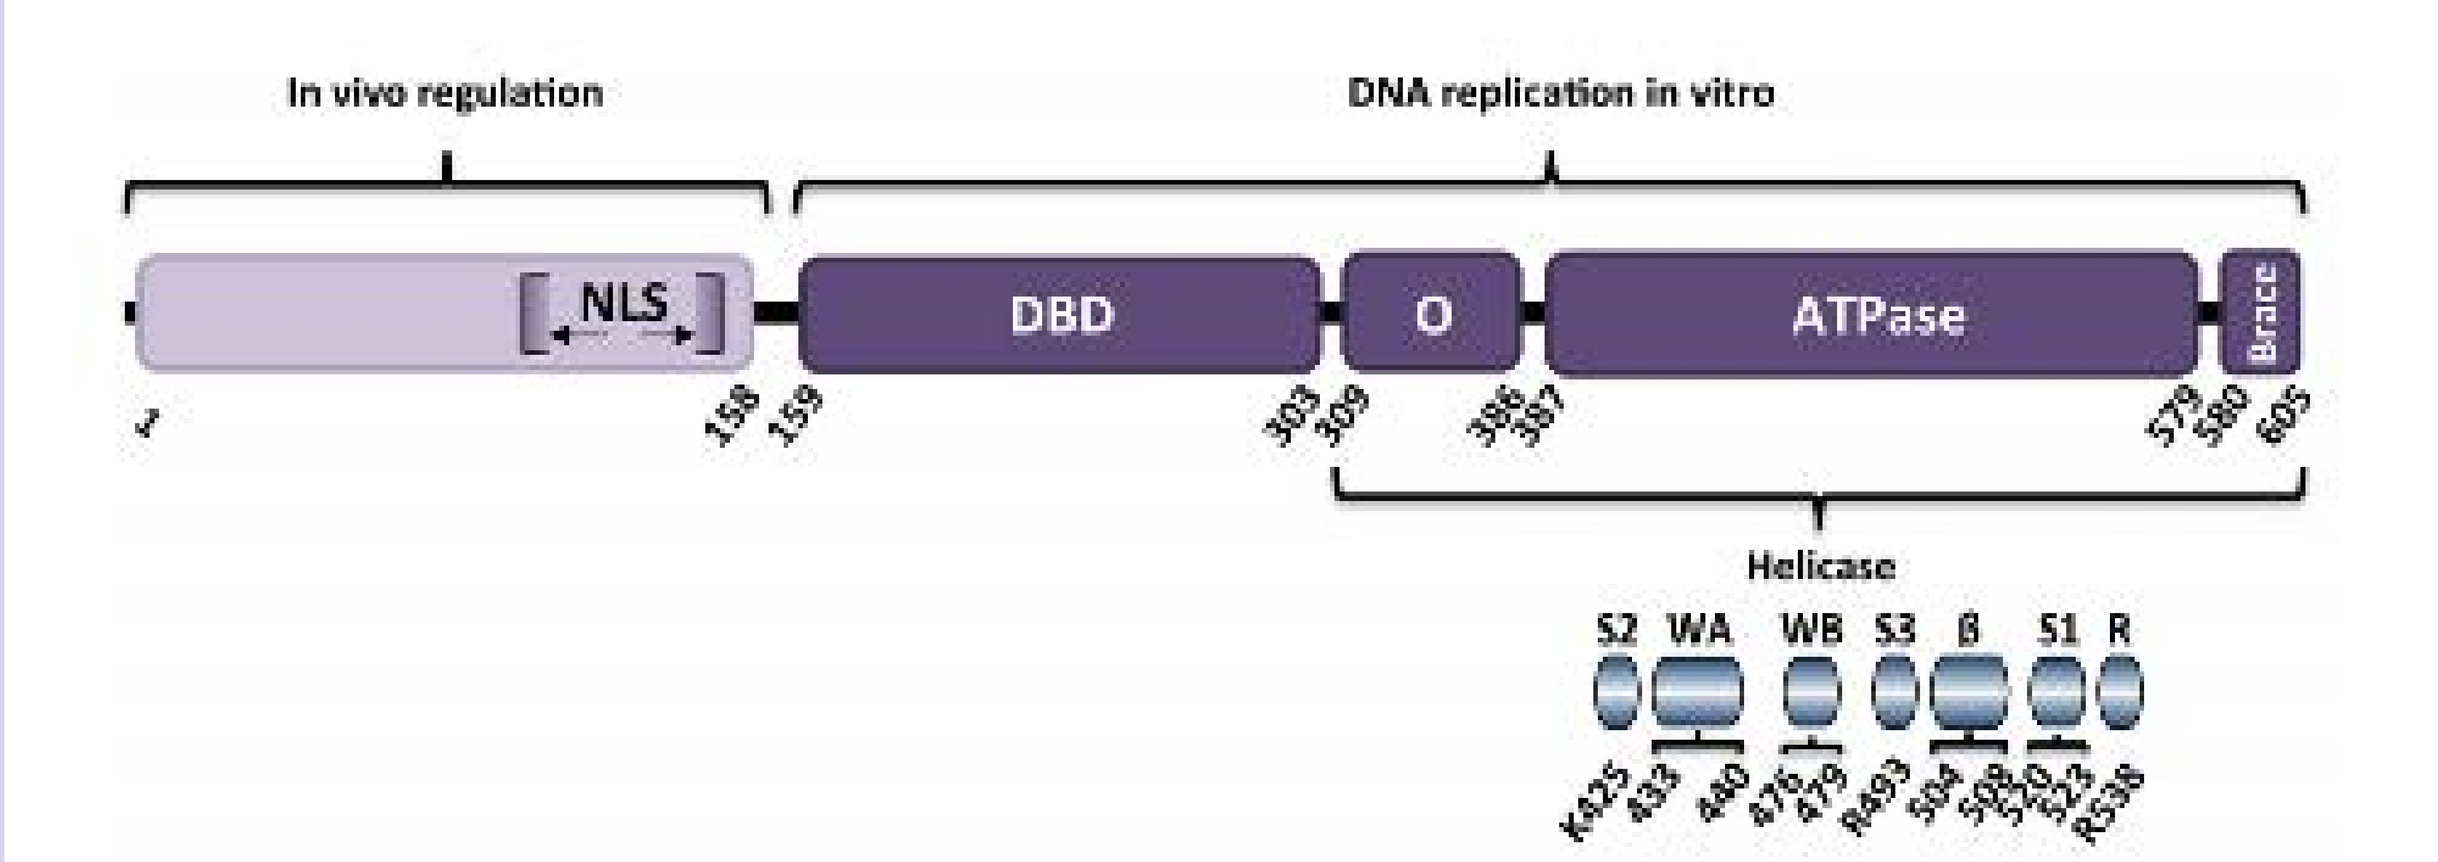
In vivo regulation
DNA replication in vitro
NLS
DBD
O
ATPase
Brace
1
158
159
303
309
386
387
579
580
605
Helicase
S2 WA WB S3 β S1 R
K425 433 440 476 479 R493 504 508 520 523 R538

Modeling the structure of the E1 helicase molecular machine leads to a deeper understanding of how it functions. The asymmetric rotary model describes the pattern of conformational changes in the subunits, which rotate from a single ATP state, to four intermediate dephosphorylated states, to a single apo (or empty) state. The ATP-type state, is characterized by the beta-hairpin in its highest position reaching up the ssDNA strand to bind to a γ-phosphate farther down the chain. The ATP-like coordination has multiple interactions within the ATP binding site. Sensor 2 K425 engages the α-phosphate of ATP with tyrosine D499 in addition to engaging the β-Phosphate. Sensor 3 binds to the Walker B aspartate and Sensor 1 as well as the nearby D489. These interactions hold ATP in place as the catalytic work is carried out by the arginine finger R538 and Sensor 1 which engage the γ-Phosphate of the ATP.The ADP-type bound state corresponds with the intermediate, middle position of the β-hairpin. This configuration has fewer interactions than the ATP-type. Sensor 3 has interactions with Walker B and Sensor 1 as in the previous state. Y534 of the arginine finger is sandwiched between R538 of the arginine finger and sensor 3, preventing interactions observed in the ATP-like state. The location of K425 Sensor 2 is too distant to engage ADP while Walker A remains bound to the β-phosphate of the ADP and Y499 interacts with the α-phosphate.The Apo state results in the β-hairpin in the lowest position in the central channel. There are no observed interactions in the active site at this conformation as there is no ATP bound and no interactions between monomers.

ATP active site with ADP

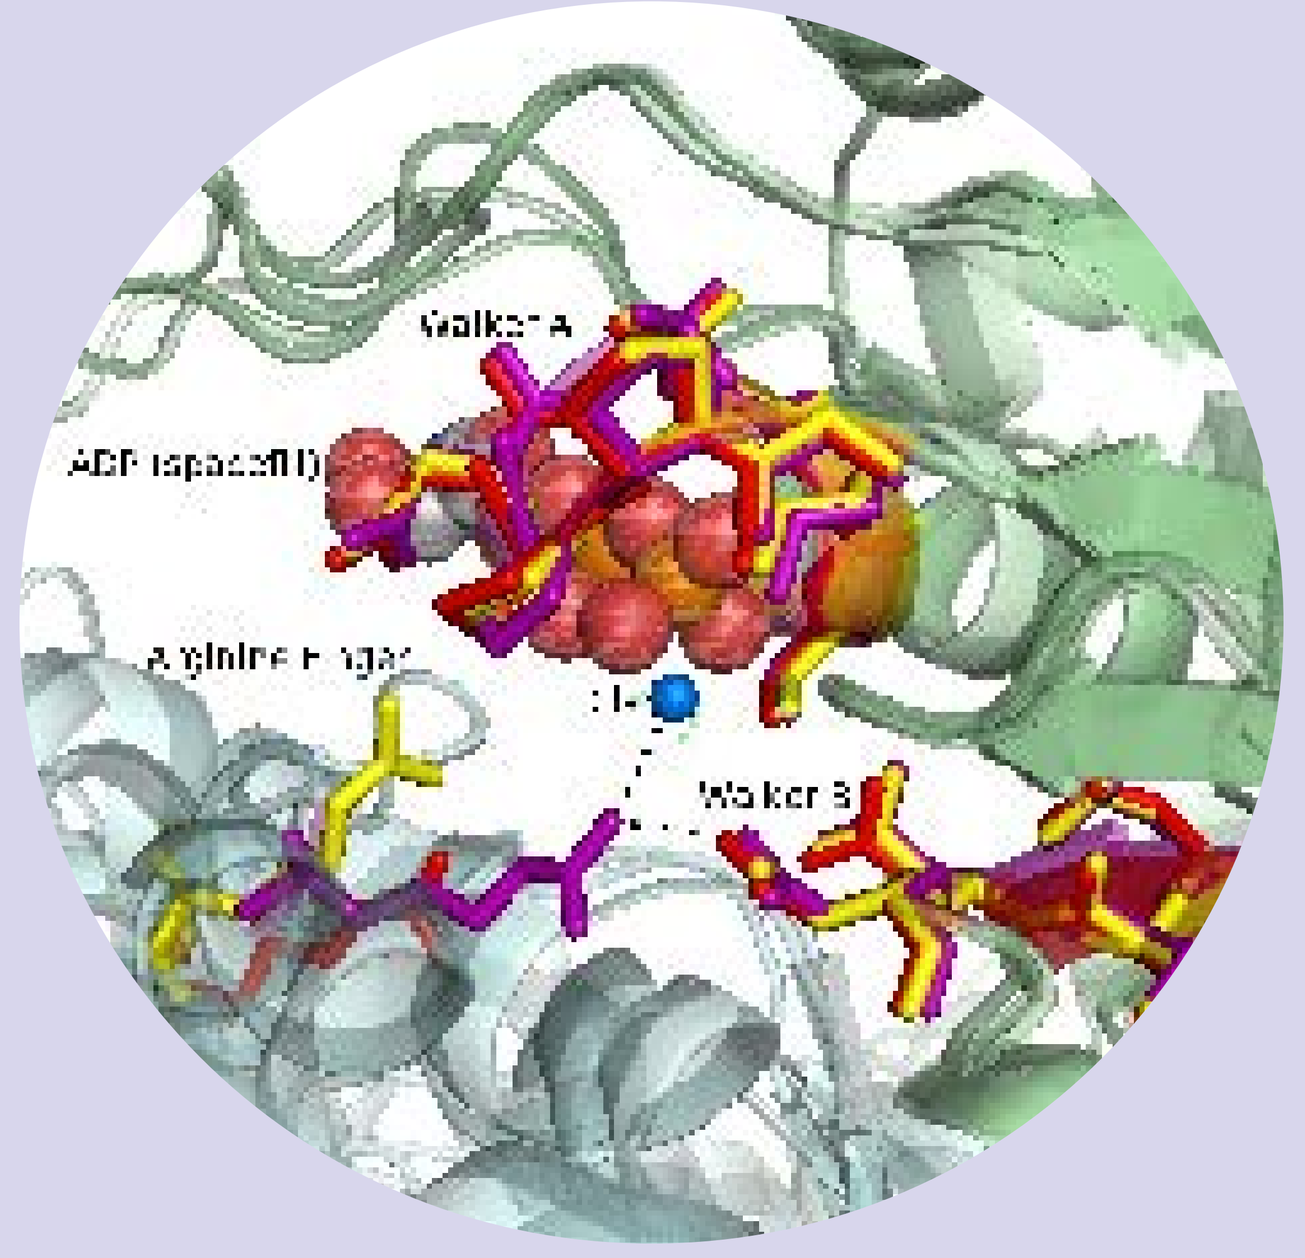
Walker A
Walker B

Sticks color-coded by state
Red - apo, purple - ATP tightly bound, yellow - ADP

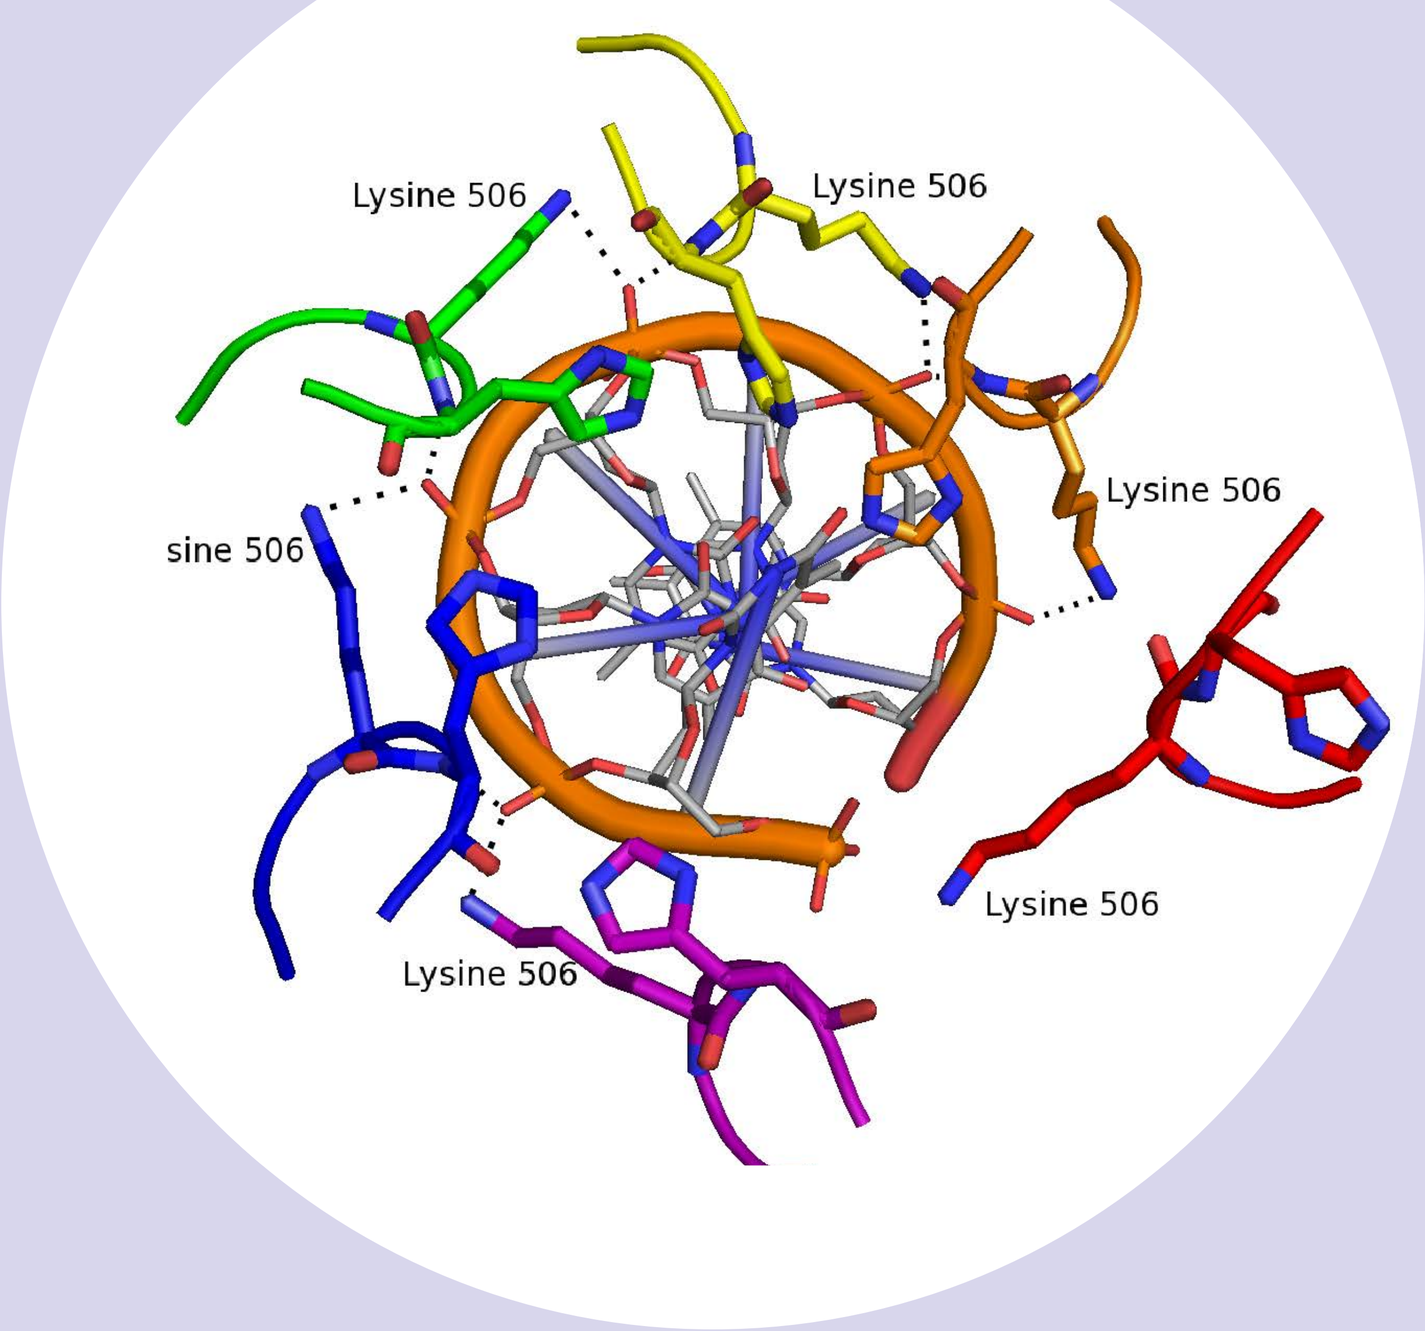
Lysine 506
Lysine 506
Lysine 506
sine 506
Lysine 506
Lysine 506

β-hairpin interactions with DNA (Pymol PDB ID: 2GXA)

Proposed ATP hydrolysis models

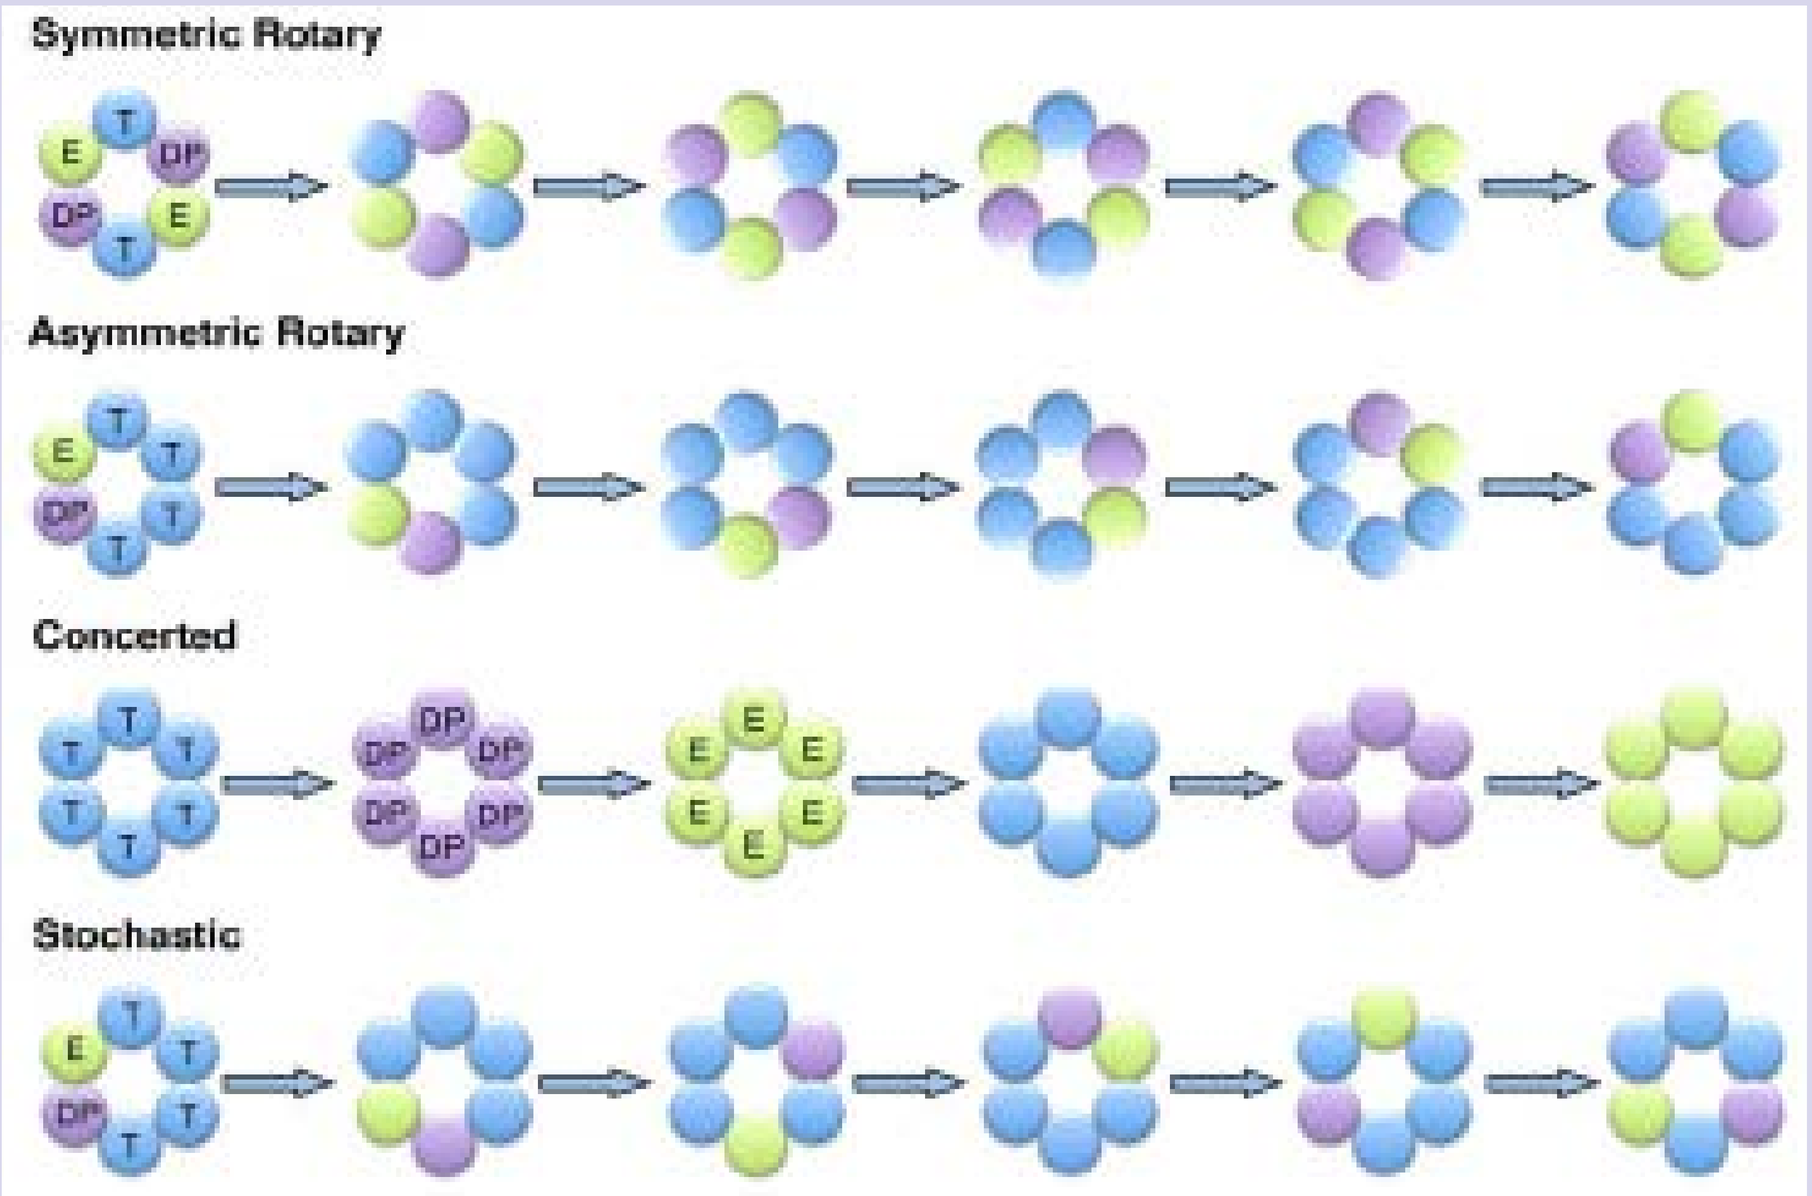
Symmetric Rotary
Asymmetric Rotary
Concerted
Stochastic

β-hairpin conformation changes. (Pymol PDB ID: 2GXA)

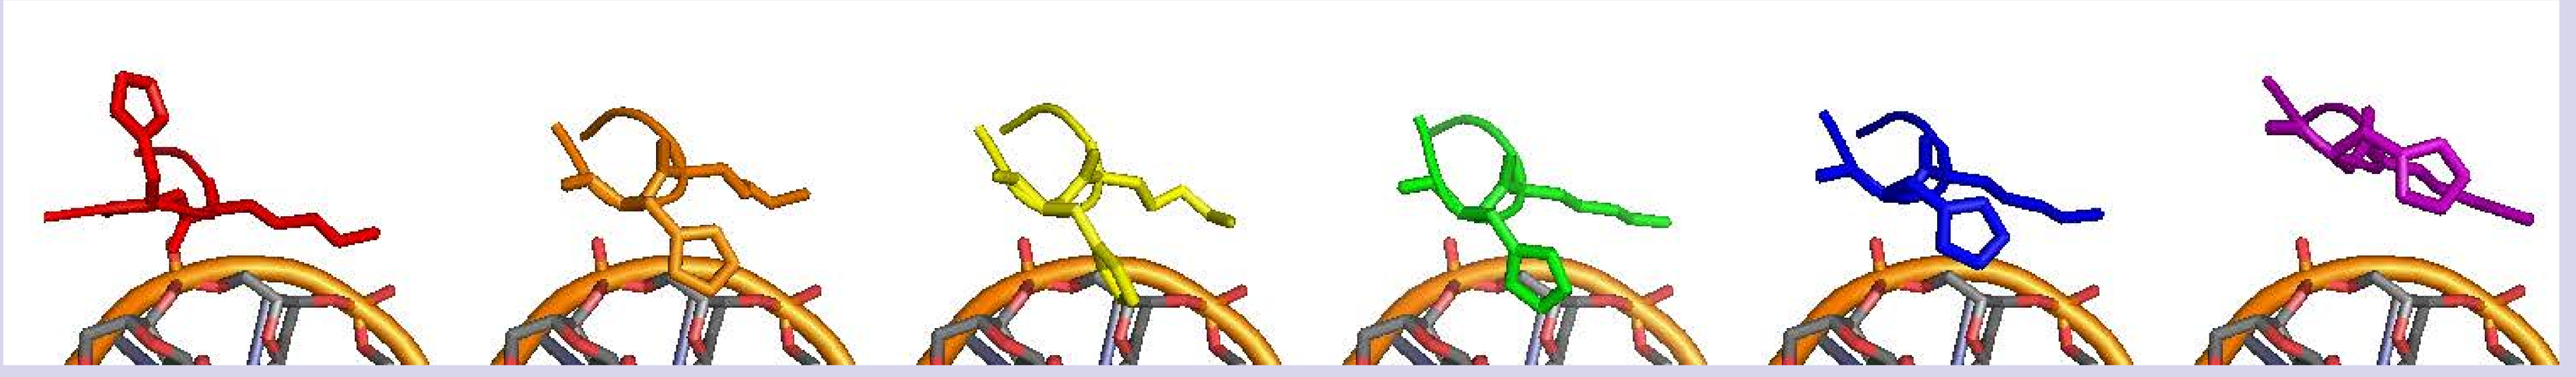

Works cited:
Monika Bergvall, Thomas Melendy, Jacques Archambault (2013). The E1 Proteins. Virology, 445:36 Monika Bergvall, Thomas Melendy, Jacques Archambault (2013). The E1 Proteins. Virology, 445:42; Eric J. Enemark, Leemor Joshua-Tor (2006). Mechanism of DNA translocation in a replicative hexameric helicase. Nature, 442:270-275; Artem Y Lyubimov, Melania Strycharska, James M Berger (2011). The Nuts and Bolts of Ring-Translocase Structure and Mechanism. Current Opinion in Structural Biology, 21:243; Ciaran B. J. Woodman, Stuart I. Collins & Lawrence S. Young (2007). The natural history of cervical HPV infection: unresolved issues. Nature Reviews Cancer, 7:11-22; Smita S Patel, Manjula Pandey, Divya Nandakumar (2011). Dynamic coupling between the motors of DNA replication: hexameric helicase, DNA polymerase, and primase. Current Opinion in Chemical Biology 15: 5; Monika Bergvall, Thomas Melendy, Jacque Archambault(2013). The E1 Proteins. Virology 445:35-56; Smita S Patel, Manjula Pandey, Divya Nandakmar(2011). Dynamic coupling between the motors of DNA replication: hexameric helicase, DNA polymerase, and primase. Current Opinion in Chemical Biology 15:1–11; Artem Y Lyubimov, Melania Strycharska, James M berger(2011). The nuts and bolts of ring-translocase structure and mechanism. Current Opinion in Structural Biology 21:240-249